Ozone Detection in Pharmaceutical Containers

Charlotte Bell[a], Elaine Martin[a], Brendan Fish[b]
Biopharmaceutical and Bioprocessing Technology Centre, Newcastle University, UK
GlaxoSmithKline, Barnard Castle, UK
charlotte.bell3@ncl.ac.uk

1 Introduction

Pharmaceutical vials are subjected to a high voltage leak detection (HVLD) test. A high voltage (up to 30kV) is passed across the surface of the vial and the presence of a crack is detected through a change in the current. HVLD has been linked to the breakdown of active pharmaceutical ingredients (APIs) and it is hypothesised that ozone, a strong oxidising agent, generated as a consequence of the high voltage, is responsible. (1) It is crucial that the manufacturing process is designed to minimise any changes to the molecular structure of biopharmaceuticals and this extends to ensuring that container integrity testing does not cause product degradation.

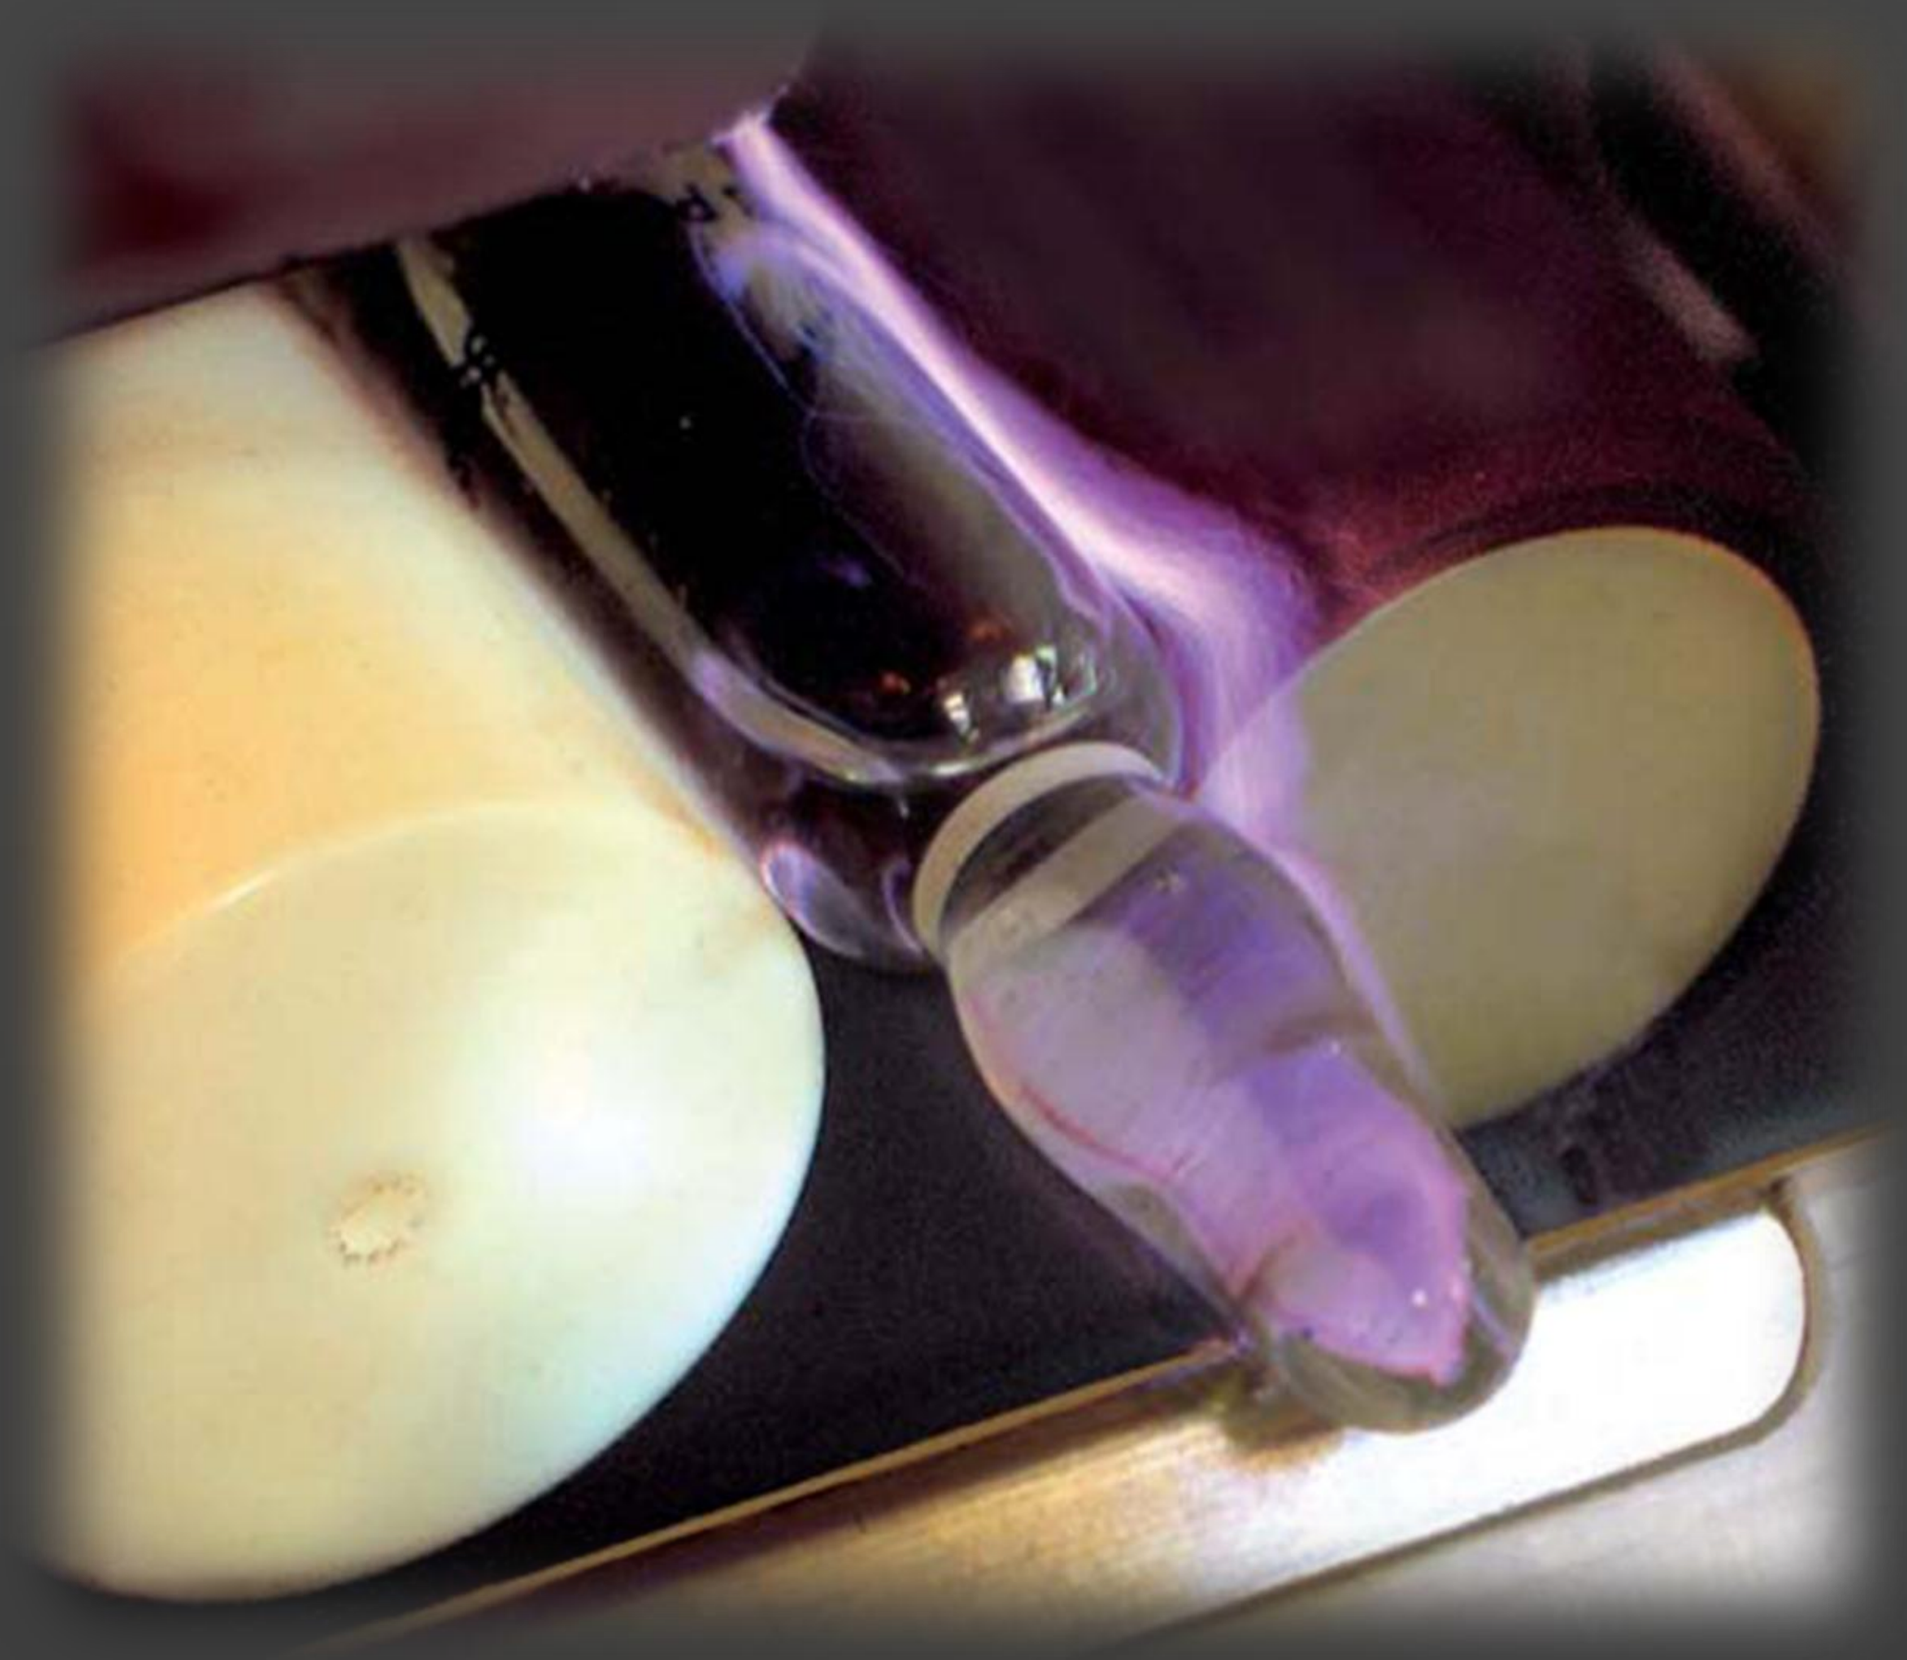

Figure 1 – Ampoule going through HVLD machine

2 Action of Ozone on Proteins

Concern exists surrounding the use of HVLD for biopharmaceuticals due to the possible degradation caused by the ozone attacking the proteins. Figure 2 shows the effect of ozone on histidine and methionine. Previous studies (2, 3) have shown the following:

- Ozone attack is generally directed towards aromatic rings
- Amide bonds of the main protein chain are not degraded by ozone, so no significant chain scission occurs and the primary protein structure remains intact
- Ozone is found to cause changes in secondary and tertiary protein structures, due to the partial oxidation of aromatic monomeric units and/or cysteine units
- Thiol groups in cysteine form disulphide cross links, causing denaturation and changes in solubility that could lead to precipitation
- Histidine displays reactivity to ozone, due to the presence of an imidazole ring, which is in agreement with the Hospira study (1) when oxidation of the imidazole ring in the drug product occurred

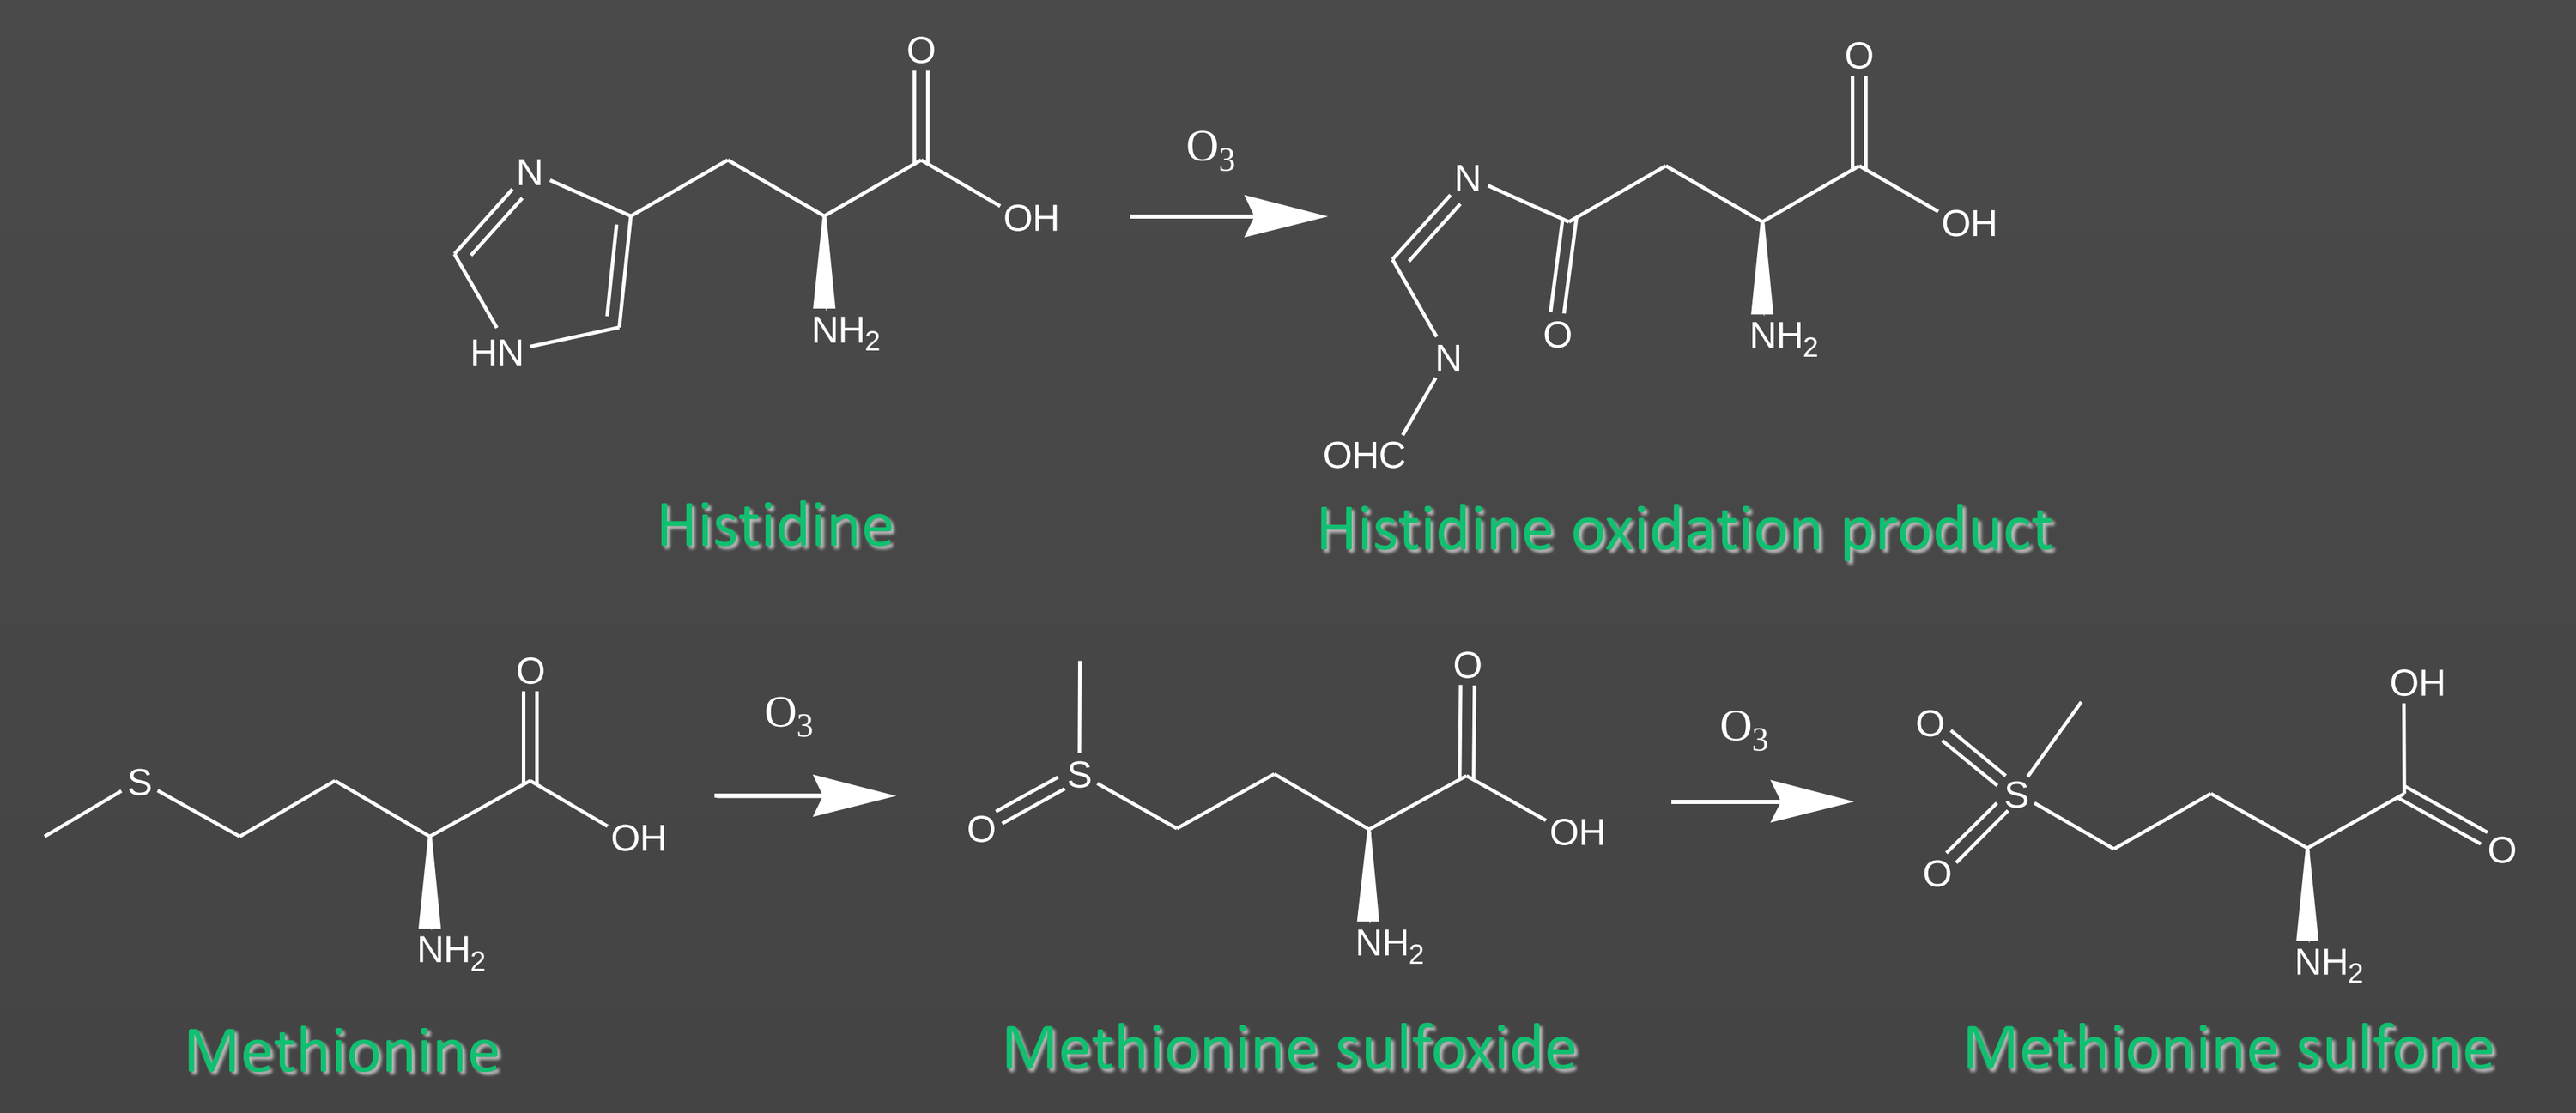
Histidine
Histidine oxidation product
Methionine
Methionine sulfoxide
Methionine sulfone

Figure 2 – Effect of ozone on histidine and methionine

3 Aims

(i) To find out if the HVLD machine causes the formation of ozone inside a vial
(ii) How can ozone inside the vial can be quantified?

The approach that will be taken is to:
1. Develop an assay for ozone measurement
2. Carry out limit of detection tests
3. Perform tests on vials subjected to HVLD
4. If ozone is present, look at the impact of ozone on APIs

4 Method Development

The method must be sufficiently sensitive to measure low levels of ozone in the gaseous and aqueous phases in the vial.

Direct measurement of aqueous ozone using UV absorbance at 258nm was investigated. By adopting this approach, it was found that the low molar absorptivity and fast decomposition of ozone made it difficult to measure.

By utilising the Indigo method, the ozone present in aqueous or gaseous samples, reacts rapidly with potassium indigo trisulfonate (PIT), to form a colourless substance - sulfonated isatin, Figure 3, thereby reducing the absorbance of PIT at 600nm. (4, 5) This method rapidly identifies the presence of ozone prior to its decomposition. A variation of the Beer Lambert law ($A=\varepsilon c\ell$) is then used to calculate the ozone concentration of a sample. Gas tight syringes are used for sampling to improve accuracy and precision (6) compared to previously published methods that utilise volumetric flasks.

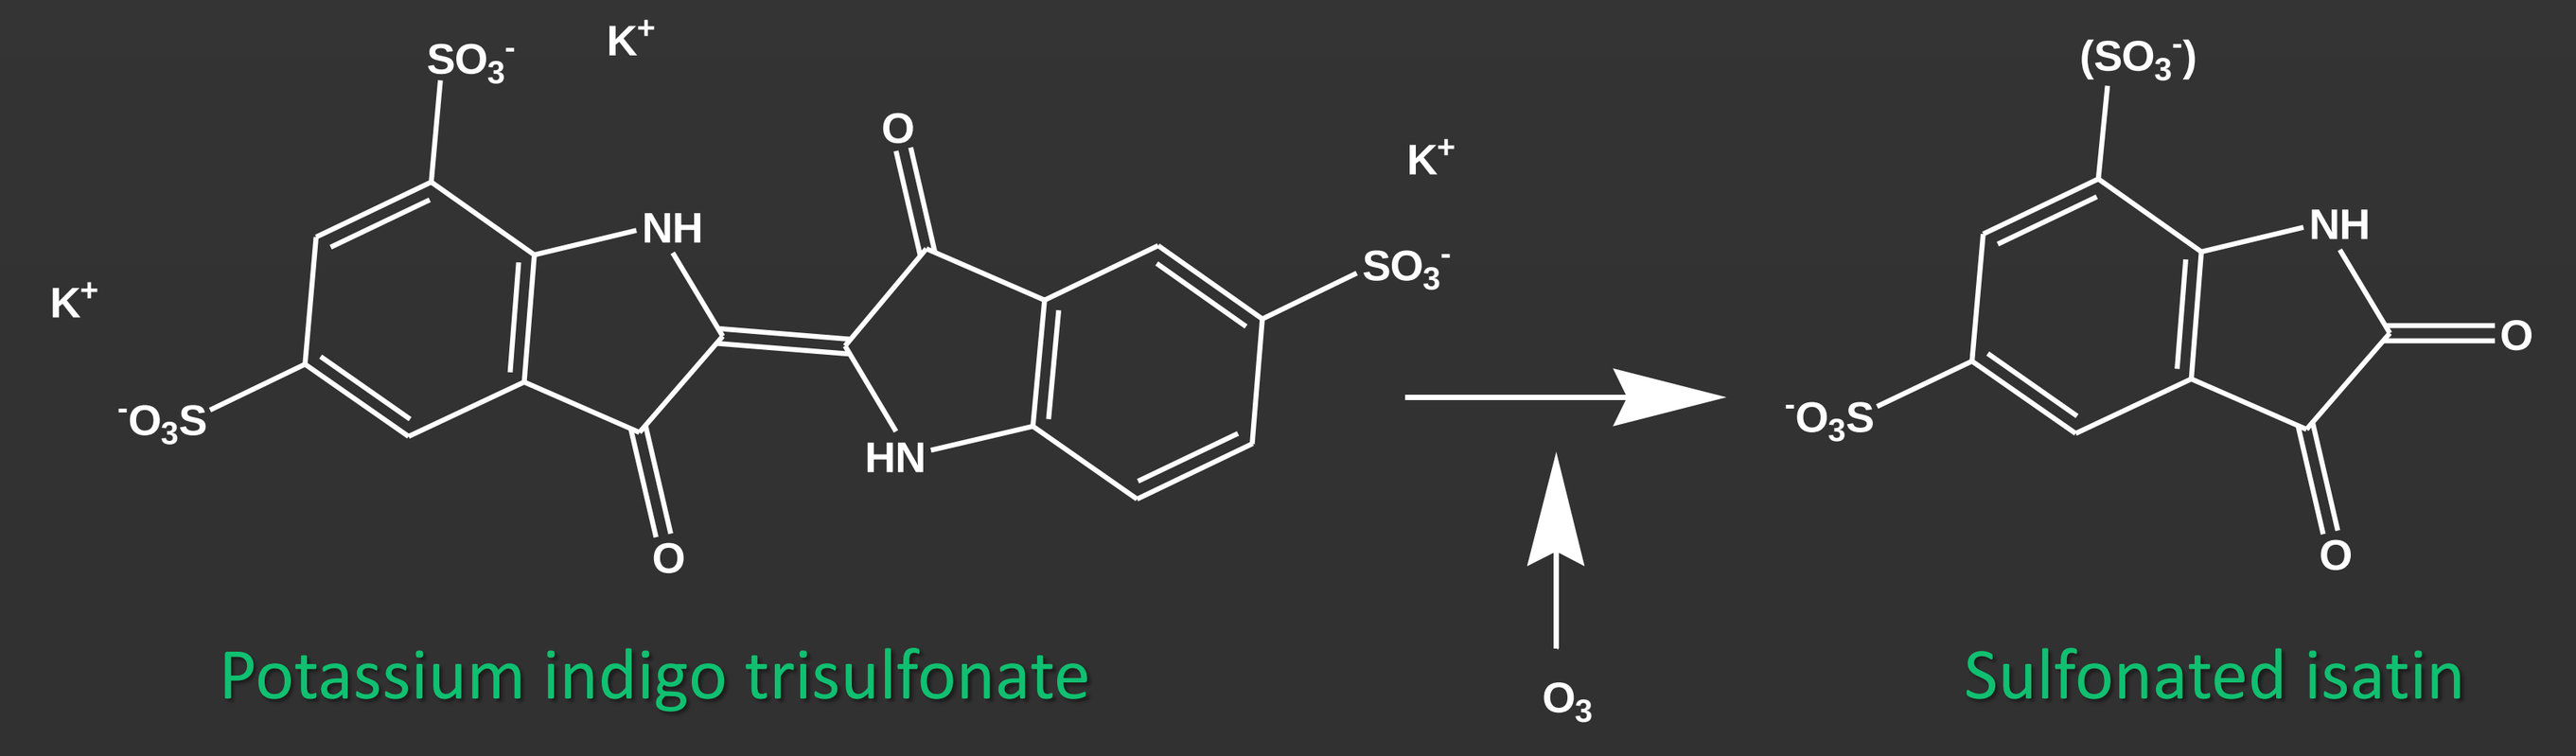
Potassium indigo trisulfonate
Sulfonated isatin

Figure 3– Conversion of PIT to colourless sulfonated isatin

5 Results and Discussion

An experiment was performed to determine how long ozone remains in solution, to understand the maximum time period available between leak testing vials with HVLD and testing for the presence of ozone. Deionised water was ozonized and the presence of ozone was measured using PIT. Figure 4 shows the decomposition of ozone over time. After only 20 minutes, the ozone initially present had decomposed, highlighting the need for a method that quickly captures the ozone after vials have been subjected to HVLD.

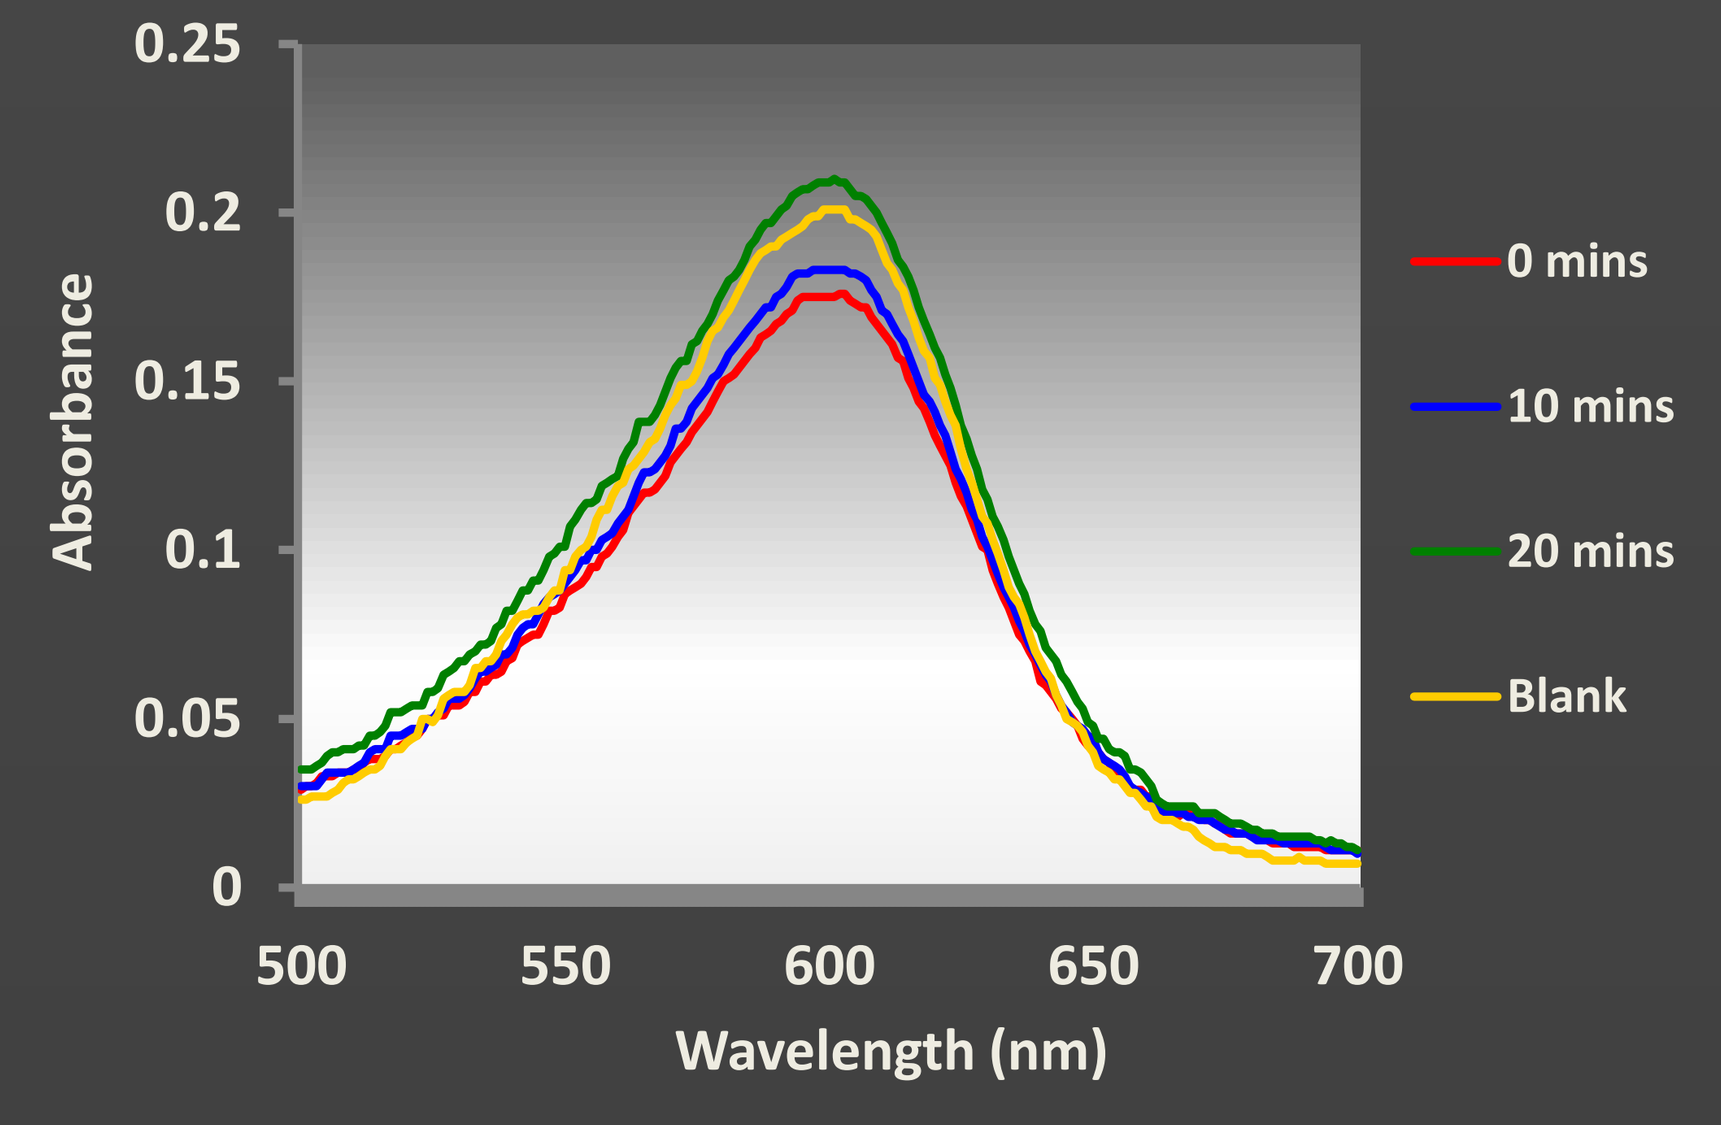
0.25
0.2
0.15
0.1
0.05
0
Absorbance
500
550
600
650
700
Wavelength (nm)
0 mins
10 mins
20 mins
Blank

Figure 4 – Ozone decomposition over time measured using PIT solution

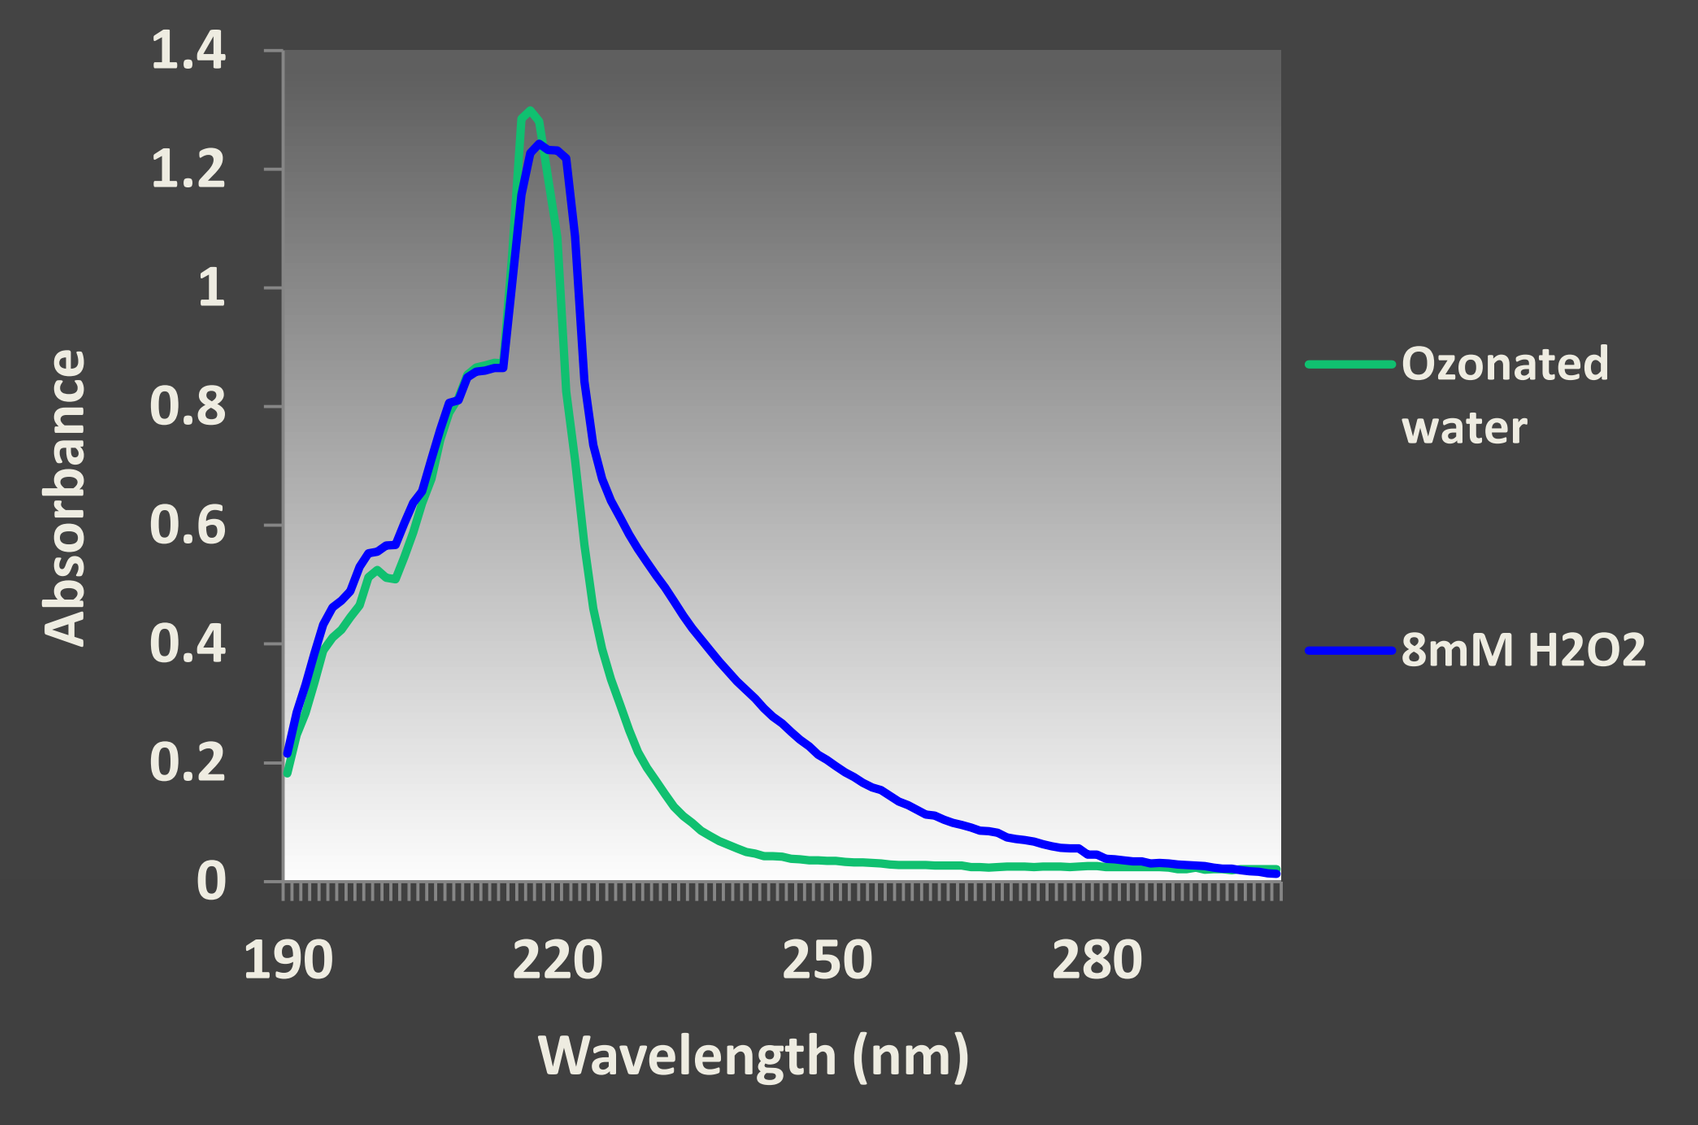
1.4
1.2
1
0.8
0.6
0.4
0.2
0
Absorbance
190
220
250
280
Wavelength (nm)
Ozonated water
8mM H2O2

Figure 5 – Presence of hydrogen peroxide in ozonized water confirmed by UV measurement

After ozonizing deionised water, the presence of hydrogen peroxide, a decomposition product of ozone, was detected at a wavelength of ~220nm. Figure 5 shows the UV spectra of ozonized water and of 8mM hydrogen peroxide. After the development of a standard curve, to relate absorbance to concentration, hydrogen peroxide could be used as an indicator to detect whether ozone was present in water, and has decomposed. Testing for hydrogen peroxide may be an important factor, as it has been previously stated that ‚*'the dissociation products of ozone in water may be more powerful oxidization agents than ozone itself'*. (7)

6 Conclusions, Future Work and Impact

Vials containing distilled water will be passed through the HVLD machine and then the headspace gas and liquid will be tested using the Indigo method. Testing for the presence of hydrogen peroxide may also give an indication that ozone has been present but has decomposed. If ozone is present in the vial, then the effects of operational parameters, e.g. voltage, on ozone generation will be investigated.

The composition of vial headspace gas could be changed to an inert gas such as nitrogen to avoid ozone generation and hence address possible product safety issues. The effect of ozone on the long-term stability of protein formulations could be investigated to determine whether protein damage occurs through contact with ozone. Currently the HVLD machine is redundant for some products. If these problems could be mitigated, the HVLD machine could be used to leak test all vials filled on the site thus increasing operational efficiency.

References
(1) McGinley, C. M., et al. Poster: Degradation of Drug Product Caused by a Leak Detection Instrument: Mechanistic Studies of Degradation. Hospira, Inc., Lake Forest, IL 60045.
(2) Cataldo, F. (2006). Ozone Degradation of Biological Macromolecules: Proteins, Hemoglobin, RNA, and DNA. Ozone: Science & Engineering **28**(5): 317-328.
(3) Sharma, V. K. and N. J. D. Graham (2010). Oxidation of Amino Acids, Peptides and Proteins by Ozone: A Review. Ozone: Science & Engineering **32**(2): 81-90.
(4) Bader H, Hoigné J (1981) Determination of ozone in water by the indigo method, Water Research 15:449-456.
(5) Bader H, Hoigné J (1982) Determination of Ozone In Water By The Indigo Method: A Submitted Standard Method, Ozone: Science & Engineering 4:169-176.
(6) Chiou C-F, Mariñas BJ, Adams JQ (1995) Modified Indigo Method For Gaseous And Aqueous Ozone Analyses, Ozone: Science & Engineering 17:329-344
(7) Peleg, M. (1976). The chemistry of ozone in the treatment of water. Water Research **10**(5): 361-365

Newcastle University · GlaxoSmithKline · EPSRC Pioneering research and skills · BBTC Biopharmaceutical Bioprocessing Technology Centre